Clinical Impact of 8% Arginine-Containing Toothpaste on Dentine Hypersensitivity

\# 1349

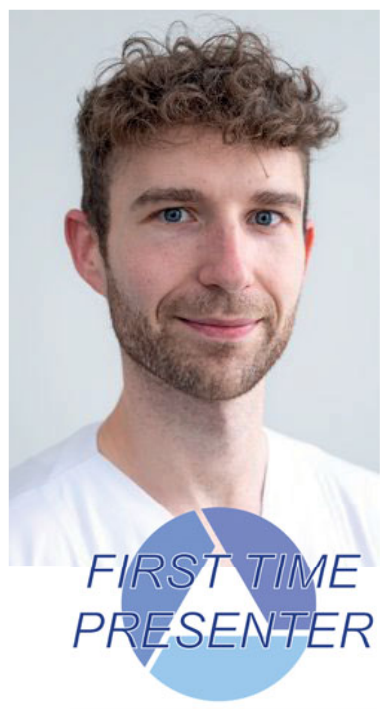
FIRST TIME
PRESENTER

C. Witting*[1], M. Michaelis[1], P.R. Schmidlin[2], T. Attin[2], K. Bitter[1], C. Heumann[3], C.R. Gernhardt[1]

[1]University Outpatient Clinic for Conservative Dentistry and Periodontology, Martin-Luther-University Halle-Wittenberg, Halle, Germany

[2]Dept. of Preventive Dentistry, Periodontology and Cariology, University of Zurich, Zurich, Switzerland

[3]Deparment of Statistics, Ludwig-Maximilians-University Munich, Munich, Germany

Introduction

Dentin hypersensitivity (DH) is a frequently observed pain condition resulting in a short sharp pain in response to thermal, evaporative, tactile, osmotic, or chemical stimuli due to the exposition of the cervical dentin surface (Fig. 1). In recent years, arginine combined with calcium carbonate was introduced and postulated to be a promising technology. In combination with calcium carbonate and phosphate, the positively charged glycoprotein arginine forms deposits on negatively charged exposed dentin surfaces, which are able to seal the open tubules and block the fluid flow mechanically leading to a sufficient pain reduction.

Objectives

The aim of this placebo-controlled, double-blinded, randomized clinical investigation was to evaluate the influence of 8% arginine-containing prophylaxis products on pain-related parameters (Schiff and VAS scores) compared to negative control in patients from a German dental clinic suffering from severe dentine hypersensitivity over 24 weeks.

Methods

After ethical approval by the Ethics Committee of the Martin Luther University Halle-Wittenberg (Germany), 101 informed patients having at least two teeth (no molars) with air blast DH scores 2, 3 (Schiff Cold Air Sensitivity Scale) were randomly assigned to either the control or test group (Fig. 2). All were treated with a single application of the in-office paste followed by daily toothpaste use (control products without desensitizing ingredients and test products containing 8% arginine, both with the same level of fluoride).The Schiff and VAS scores were evaluated before and after application of the in-office paste and after 4, 8, and 24 weeks for both study teeth (Fig. 3).

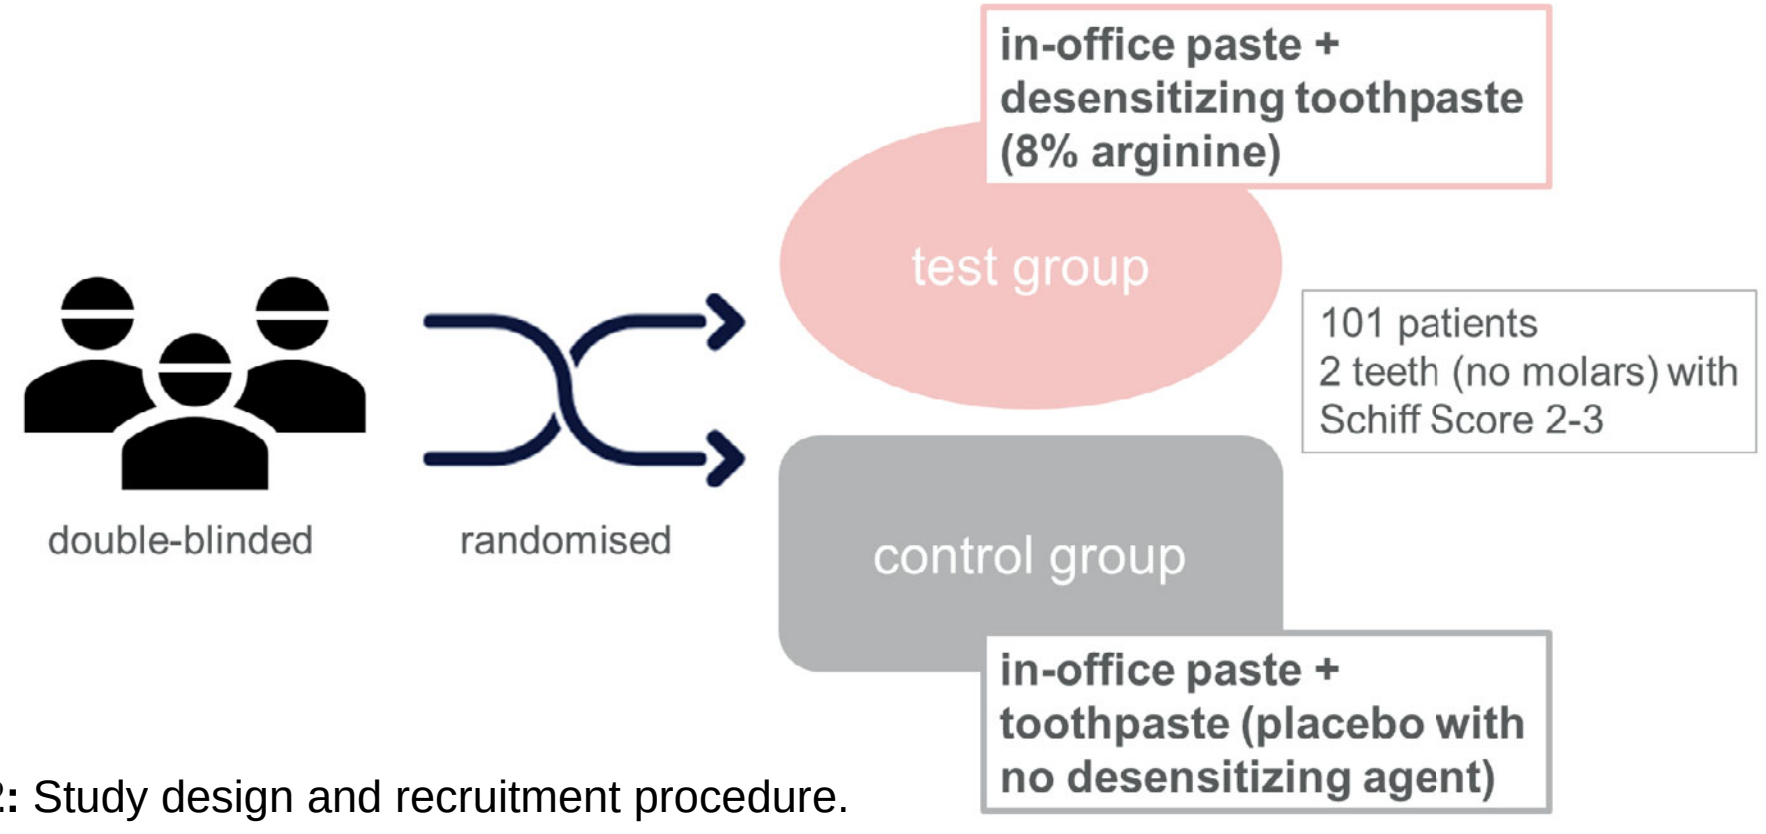
in-office paste +
desensitizing toothpaste
(8% arginine)
test group
101 patients
2 teeth (no molars) with
Schiff Score 2-3
double-blinded
randomised
control group
in-office paste +
toothpaste (placebo with
no desensitizing agent)

Fig. 2: Study design and recruitment procedure.

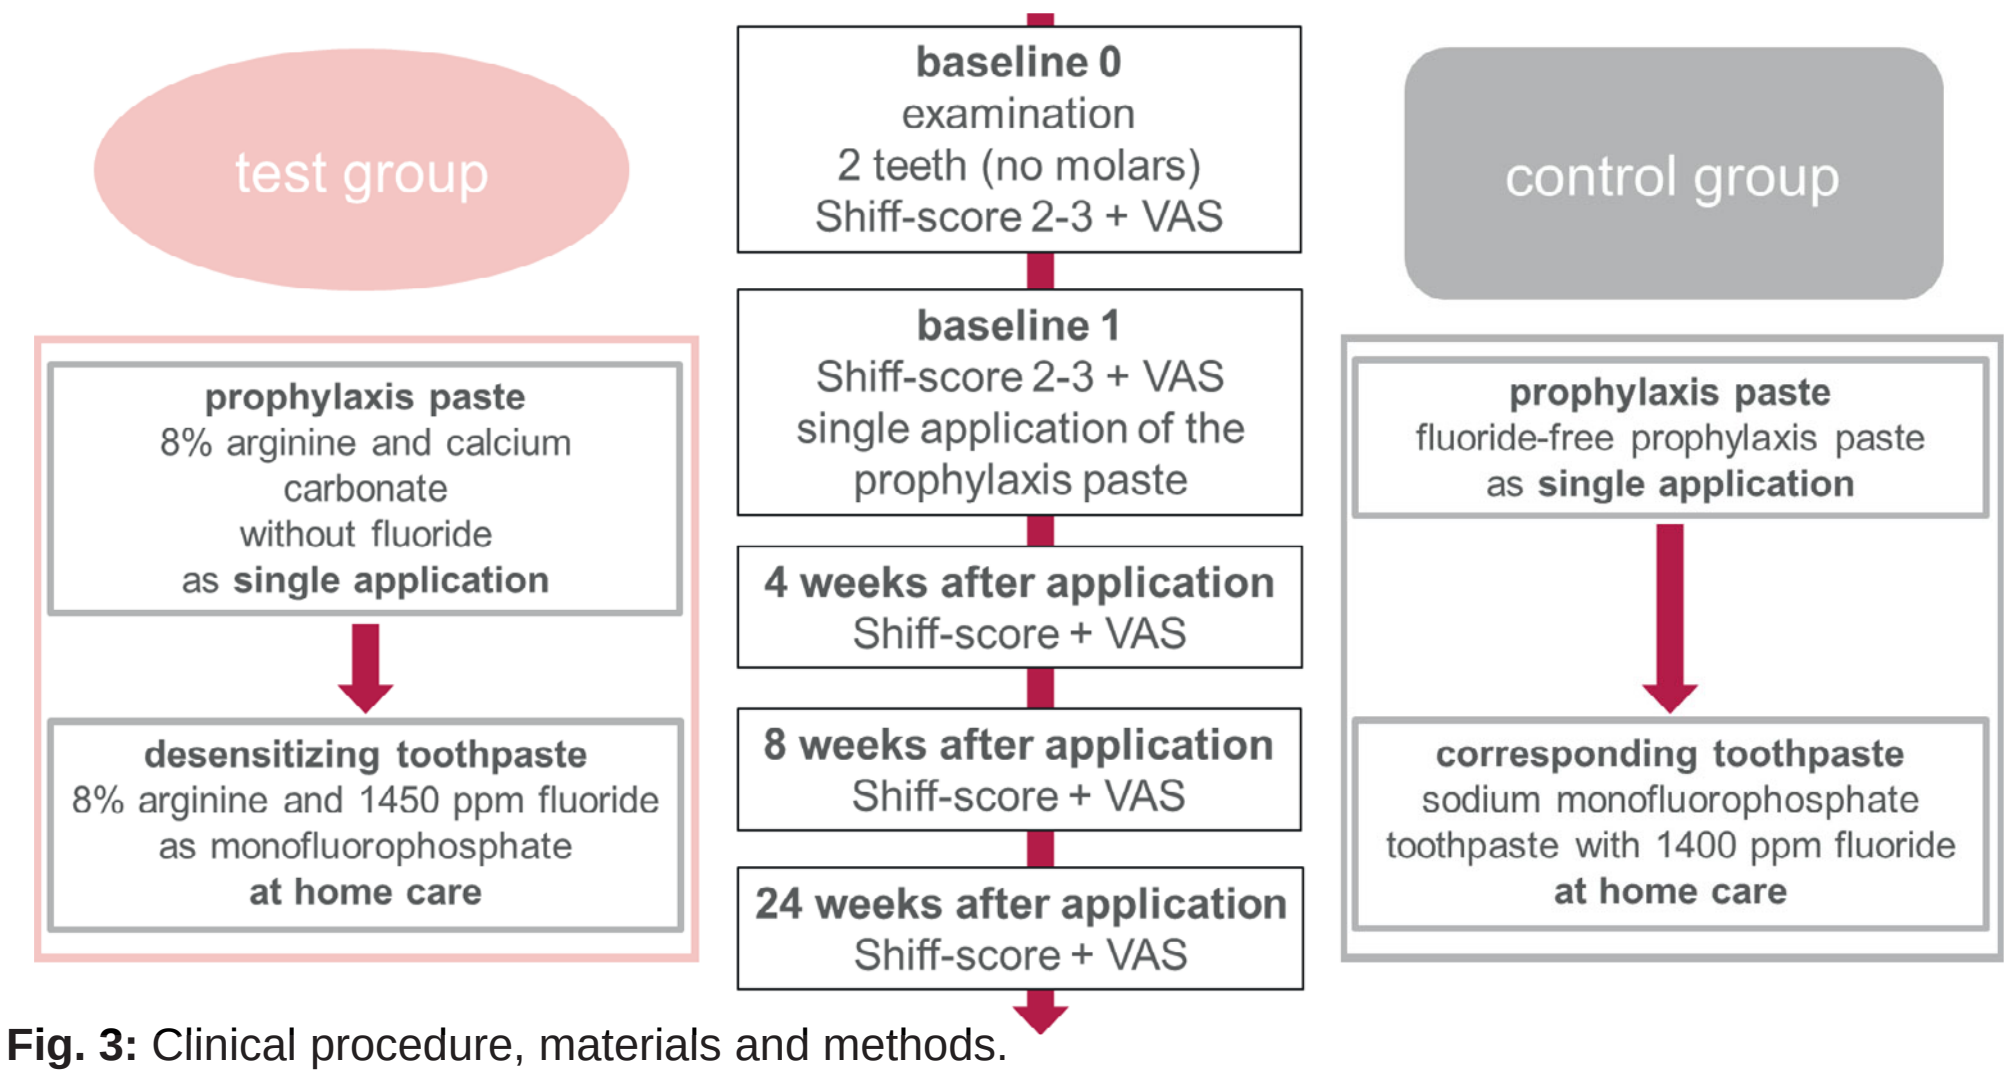
test group
baseline 0
examination
2 teeth (no molars)
Shiff-score 2-3 + VAS
control group
prophylaxis paste
8% arginine and calcium
carbonate
without fluoride
as single application
baseline 1
Shiff-score 2-3 + VAS
single application of the
prophylaxis paste
prophylaxis paste
fluoride-free prophylaxis paste
as single application
4 weeks after application
Shiff-score + VAS
8 weeks after application
Shiff-score + VAS
desensitizing toothpaste
8% arginine and 1450 ppm fluoride
as monofluorophosphate
at home care
corresponding toothpaste
sodium monofluorophosphate
toothpaste with 1400 ppm fluoride
at home care
24 weeks after application
Shiff-score + VAS

Fig. 3: Clinical procedure, materials and methods.

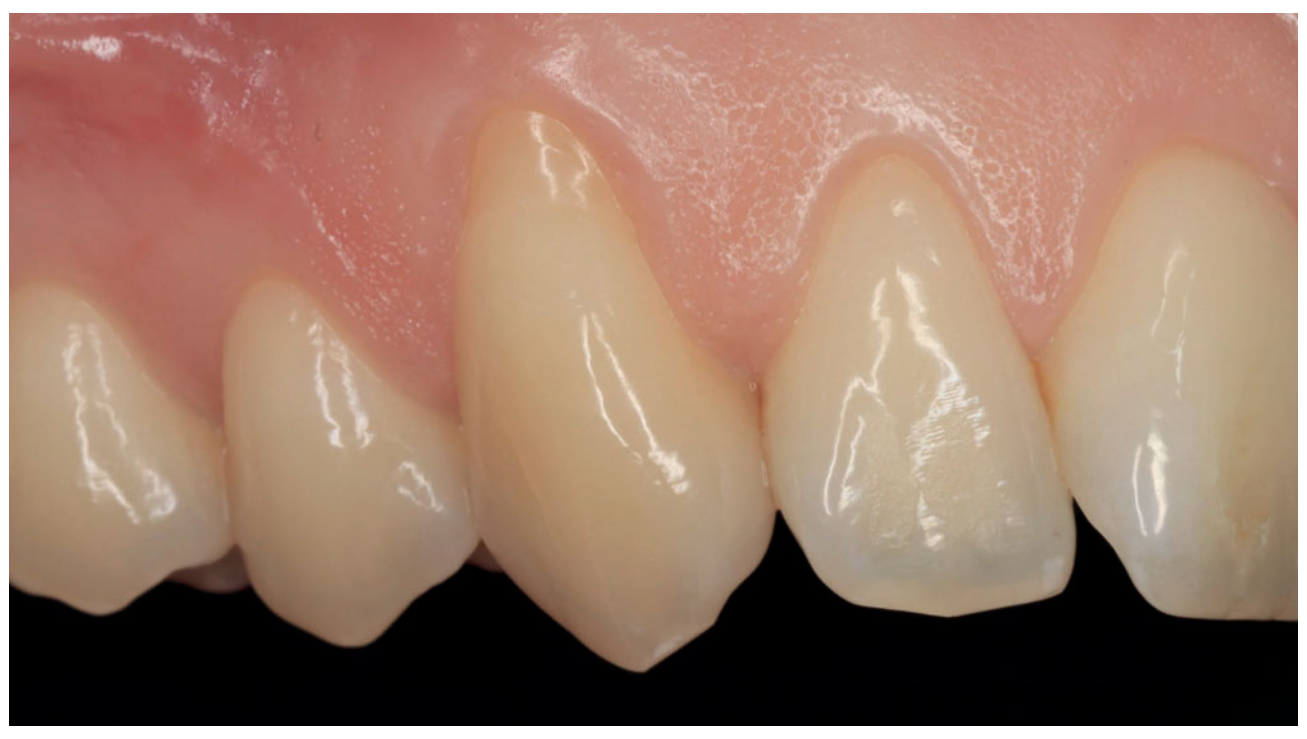

Fig. 1: A non-carious cervical lesion (NCCL) leading to dentin hypersensitivity.

Results

A total of 98 patients completed the study. The distribution of subjects with DH Schiff scores 2 and 3 at screening was similar in both groups. There were no statistically significant differences in the Schiff and VAS scores at baseline between test and control subjects (p>0.05). The pooled data for tooth 1 and tooth 2 yielded significantly greater pain relief as assessed by the Schiff score in the test group than the control group at any appointment, both in the t-test and Wilcoxon signed rank test analyses (p<0.05, Tab. 1). The VAS showed no significant differences between both groups (Tab. 2).

treatment method	evaluation time	within and intergroup analysis			
		mean ± SD (mm)		% difference between groups	
		test	control	%value	p-value
in-office treatment	Baseline 0	2.54 ± 0.42	2.51 ± 0.48	−1.2%	0.616*
	Baseline 1	1.94 ± 0.71 a)	2.29 ± 0.58 a)	15.3%	0.000**
at-home care	4-weeks	2.01 ± 0.78 b)	2.17 ± 0.68 c)	7.4%	0.030**
	8-weeks	1.78 ± 0.79 b)	1.99 ± 0.83 c)	10.6%	0.023**
	24-weeks	1.40 ± 0.90 c)	1.69 ± 0.94 c)	17.2%	0.007**

p - values for the significance of the Wilcoxon signed-rank intragroup comparison.
a) p = 0.000; statistically significant difference compared to Baseline 0.
b) p > 0.05; no statistically significant difference compared to Baseline 1.
c) p < 0.05; statistically significant difference compared to Baseline 1.
* Significance of the Ancova intergroup comparison adjusted for age and gender.
** Significance of the Ancova intergroup comparison of the Baseline 0, age and gender adjusted means

Tab. 1: Summary of the air blast sensitivity data evaluated with the Schiff score at each time point per group.

treatment method	evaluation time	within and intergroup analysis			
		mean ± SD (mm)		% difference between groups	
		test	control	%value	p-value
in-office treatment	Baseline 0	27.96 ± 25.11	25.96 ± 24.75	−7.1%	0.243*
	Baseline 1	17.69 ± 20.68 a)	19.59 ± 21.82 a)	9.7%	0.024**
at-home care	4-weeks	19.04 ± 21.77 b)	17.17 ± 18.88 b)	−9.8%	0.563**
	8-weeks	15.43 ± 19.10 b)	17.31 ± 19.71 b)	10.9%	0.219**
	24-weeks	11.89 ± 16.62 c)	14.15 ± 17.36 c)	16.0%	0.185**

p - values for the significance of the Wilcoxon signed-rank intragroup comparison.
a) p = 0.000; statistically significant difference compared to Baseline 0.
b) p > 0.05; no statistically significant difference compared to Baseline 1.
c) p < 0.05; statistically significant difference compared to Baseline 1.
* Significance of the Ancova intergroup comparison adjusted for age and gender.
** Significance of the Ancova intergroup comparison of the Baseline 0, age and gender adjusted means

Tab. 2: Summary of the tactile sensitivity data evaluated with the Visual Analog Scale (VAS) in mm at each time point per group

Conclusions

Significant improvements (Schiff score, pain relief) in a German population were demonstrated after application of the in-office paste and over the 24-week period of brushing with the 8.0% arginine-containing toothpaste.

This study was supported by Colgate-Palmolive Europe, Therwil, Switzerland.

102nd General Session & Exhibition of the IADR
53rd Annual Meeting of the AADOCR
48th Annual Meeting of the CADR
New Orleans, LA, USA, March 13-16, 2024

UNIVERSITY MEDICINE HALLE
www.umh.de

Correspondence: Carl Witting, Martin-Luther-University Halle-Wittenberg, University Outpatient Clinic for Conservative Dentistry and Periodontology, Magdeburger Strasse 16, 06112 Halle, Germany, e-Mail: carl.witting@uk-halle.de